REStoring lymphoCytes Using NKTR-255 after chemoradiothErapy in solid tumors (RESCUE): Preplanned Interim Safety and Efficacy Analysis

Steven H. Lin[1], Ying-Shiuan Chen[1], Priti Gupta[1], Madison Kahanek[1], Becky Slack Tidwell[2], Hyunsoo Hwang[2], Peter F. Thall[2], Stephen G. Chun[1], Joe Y. Chang[1], Zhongxing Liao[1], Zachary Lee[4], Mario Marcondes[4], Mary Tagliaferri[4], Jonathan Zalevsky[4], George Blumenschein[3], Janet Tu[3], John V. Heymach[3], Mehmet Altan[3].

Department of [1]Thoracic Radiation Oncology, [2]Biostatistics and [3]Thoracic Medical Oncology, MDACC; [4]Nektar Therapeutics

THE UNIVERSITY OF TEXAS MD Anderson Cancer Center
Making Cancer History®

Background

Radiation induced lymphopenia (RIL) is common after chemoradiation therapy (CRT) and can negatively impact efficacy of immunotherapies (see Figure 1). RIL has been associated with worse survival in several solid tumors, including lung, esophageal, and other cancers. Therapeutic interventions to rescue persistent RIL after CRT may confer prognostic benefits and enhance survival. IL-15 is a pleiotropic cytokine essential for lymphocyte formation, immunological memory and cell survival. NKTR-255 is an investigational polymer-conjugated rhIL-15 receptor agonist with extended exposure and sustained activation of the IL-15 pathway. Here, we present a prespecified interim analysis of a Phase II study evaluating safety and PK of NKTR-255 for restoring absolute lymphocyte count (ALC) after CRT in locally advanced non-small cell lung cancer (LA-NSCLC) patients.

Figure 1. Severe Radiation-Induced Lymphopenia Attenuates Benefits of Durvalumab after Concurrent Chemoradiation for Stage 3 NSCLC[3,4]

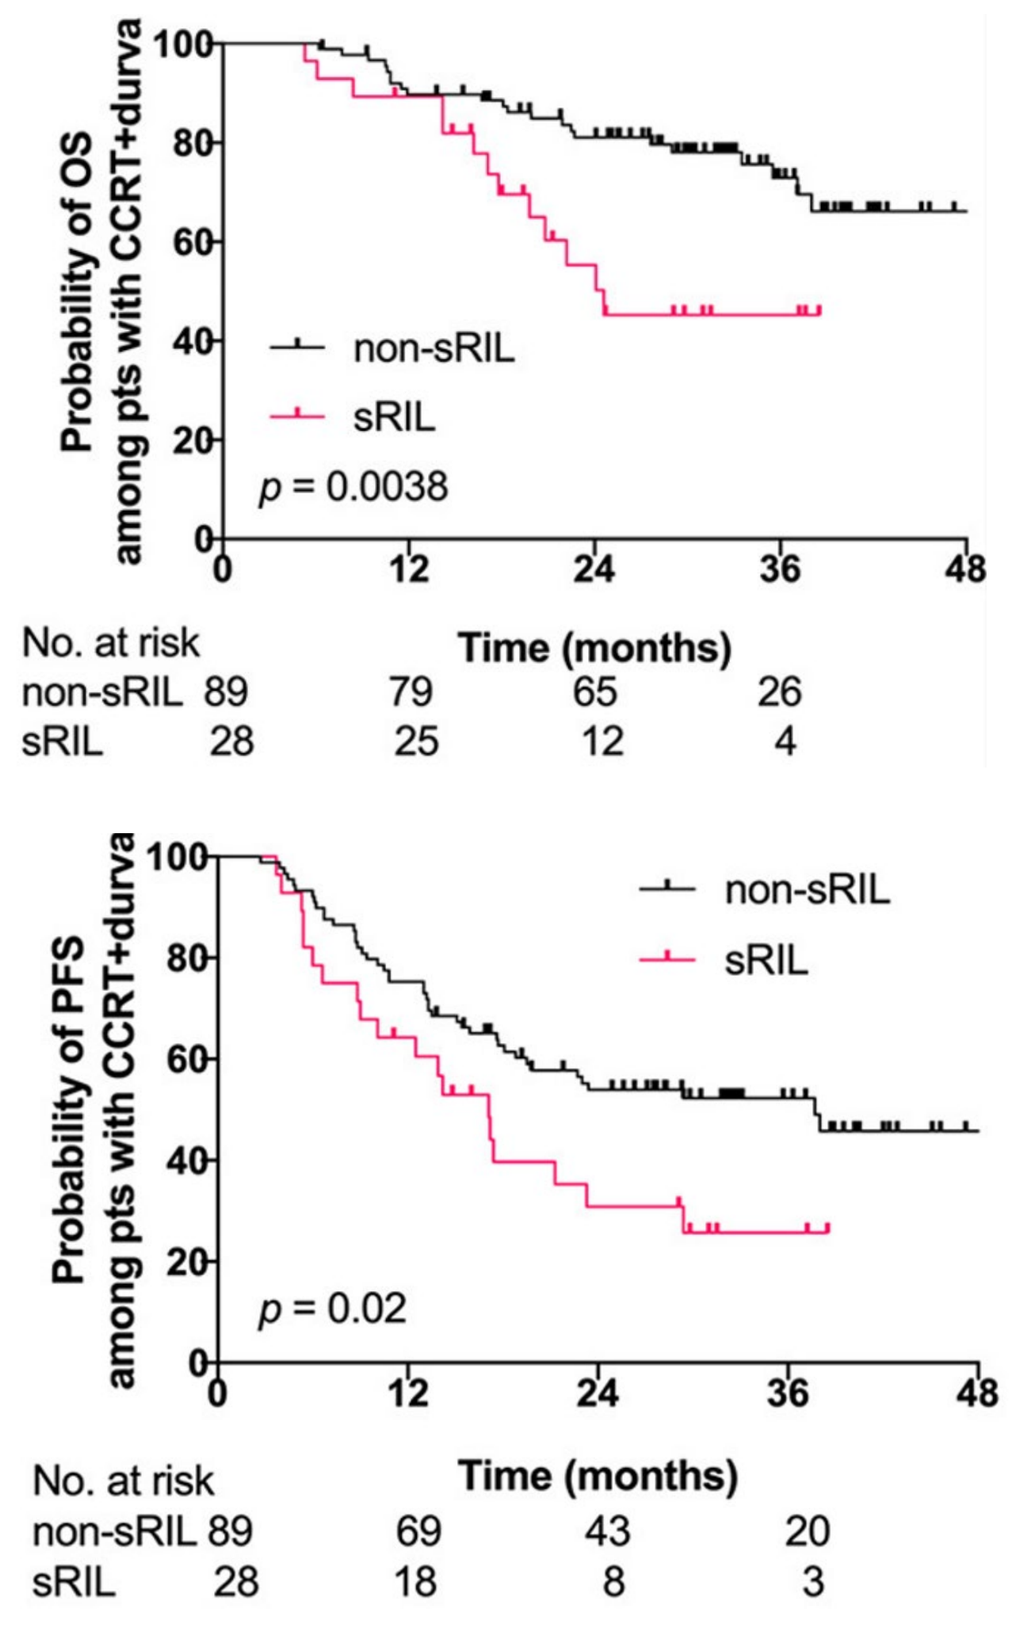
Probability of OS among pts with CCRT+durva
non-sRIL
sRIL
p = 0.0038
No. at risk
Time (months)
non-sRIL 89 79 65 26
sRIL 28 25 12 4
Probability of PFS among pts with CCRT+durva
p = 0.02
non-sRIL 89 69 43 20
sRIL 28 18 8 3

Methods

NKTR-255 (3µg/kg IV) is administered after CRT (day 0), with repeat dosing every 4 weeks concurrently with durvalumab (1500mg IV) up to 1 year. The primary objectives are safety, incidence of treatment-related adverse events (TRAE) Grade 3+ (G3) radiation pneumonitis, and ALC normalization after NKTR-255 and durvalumab at Week 8 after CRT. Early stopping rules are in place to control incidence of G3+ radiation pneumonitis, and futility monitoring for efficacy, defined as ALC recovery >1000 cells/µL.

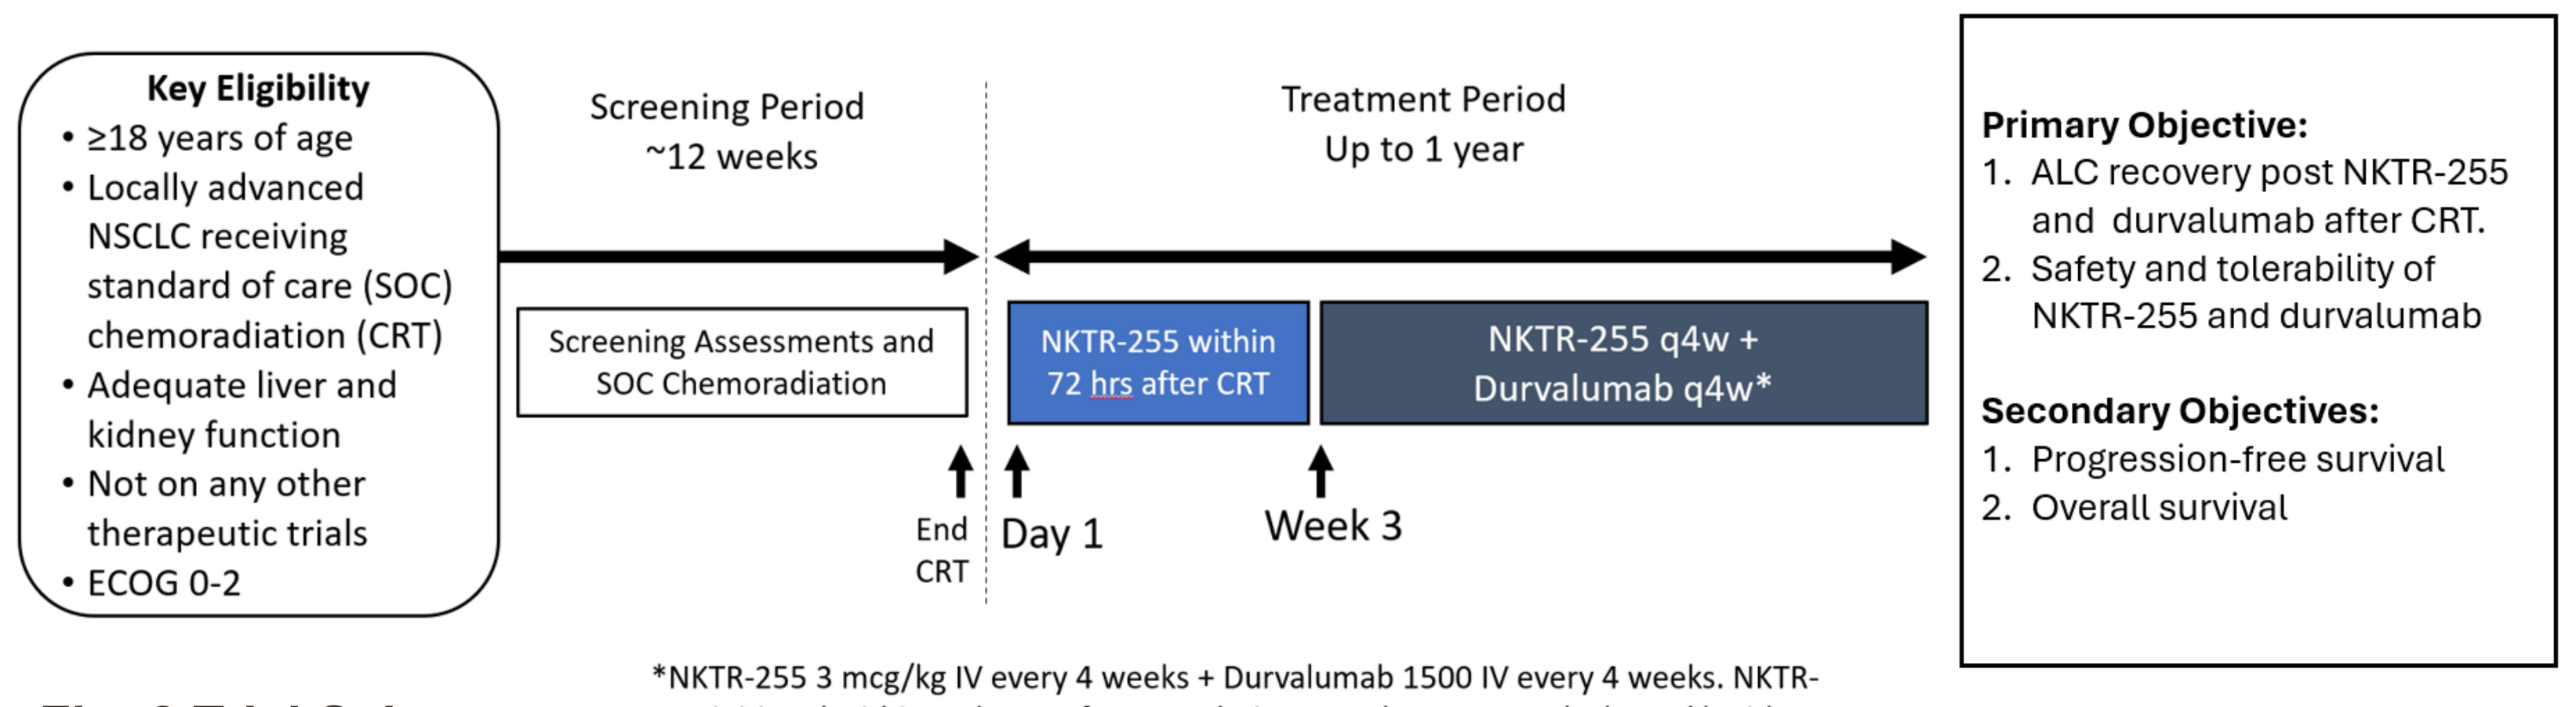
Key Eligibility
• ≥18 years of age
• Locally advanced NSCLC receiving standard of care (SOC) chemoradiation (CRT)
• Adequate liver and kidney function
• Not on any other therapeutic trials
• ECOG 0-2
Screening Period ~12 weeks
Treatment Period Up to 1 year
Screening Assessments and SOC Chemoradiation
NKTR-255 within 72 hrs after CRT
NKTR-255 q4w + Durvalumab q4w*
End CRT
Day 1
Week 3
Primary Objective:
1. ALC recovery post NKTR-255 and durvalumab after CRT.
2. Safety and tolerability of NKTR-255 and durvalumab
Secondary Objectives:
1. Progression-free survival
2. Overall survival

Fig. 2 Trial Schema

*NKTR-255 3 mcg/kg IV every 4 weeks + Durvalumab 1500 IV every 4 weeks. NKTR-255 initiated within 72 hours after completing CRT, then at 3 weeks (± 1 wk) with durvalumab and then every 4 weeks (± 1 wk) with durvalumab for up to 1 year

Results

As of October 15th, 2024, 15 patients received ≥ 1 dose of NKTR-255 (median 5 cycles, with a median time on treatment of 4.2 months). The most common grade 1-2 TRAEs were fever (n=8), chills (n=6), dyspnea (n=6), sinus tachycardia (n=5), and fatigue (n=5). One patient developed G3 pneumonitis. Two patients had transient G3 liver enzyme elevation and 1 patient had transient G3 sinus tachycardia. Among the 15 patients, 10 patients completed ≥2 doses of study treatment and 9 patients reached week 8 ALC evaluation. 1 patient received 2 doses of study treatment but had to receive salvage therapy due to progressive disease prior to week 8.

Table. 1 Adverse Events observed from at least 2 patients Related to Durvalumab or NKTR-255

TRAES by Max Grade	N	%	G1 Mild	G2 Mod.	G3 Sev.
Fever	8	53%	8	-	-
Chills	6	40%	6	-	-
Dyspnea	6	40%	1	5	-
Fatigue	5	33%	1	4	-
Sinus Tachycardia	5	33%	3	1	1
Cough	4	27%	4	-	-
Rash Maculo-Papular	4	27%	4	-	-
ALT Increased	3	20%	1	1	1
Diarrhea	3	20%	3	-	-
Flu Like Symptoms	3	20%	2	1	-
Anorexia	2	13%	2	-	-
AST Increased	2	13%	-	2	-
Dizziness	2	13%	2	-	-
Pneumonitis	2	13%	-	1	1
Vomiting	2	13%	2	-	-

NKTR-255 Rescues CRT-induced Lymphopenia Post-Chemoradiation (CRT)

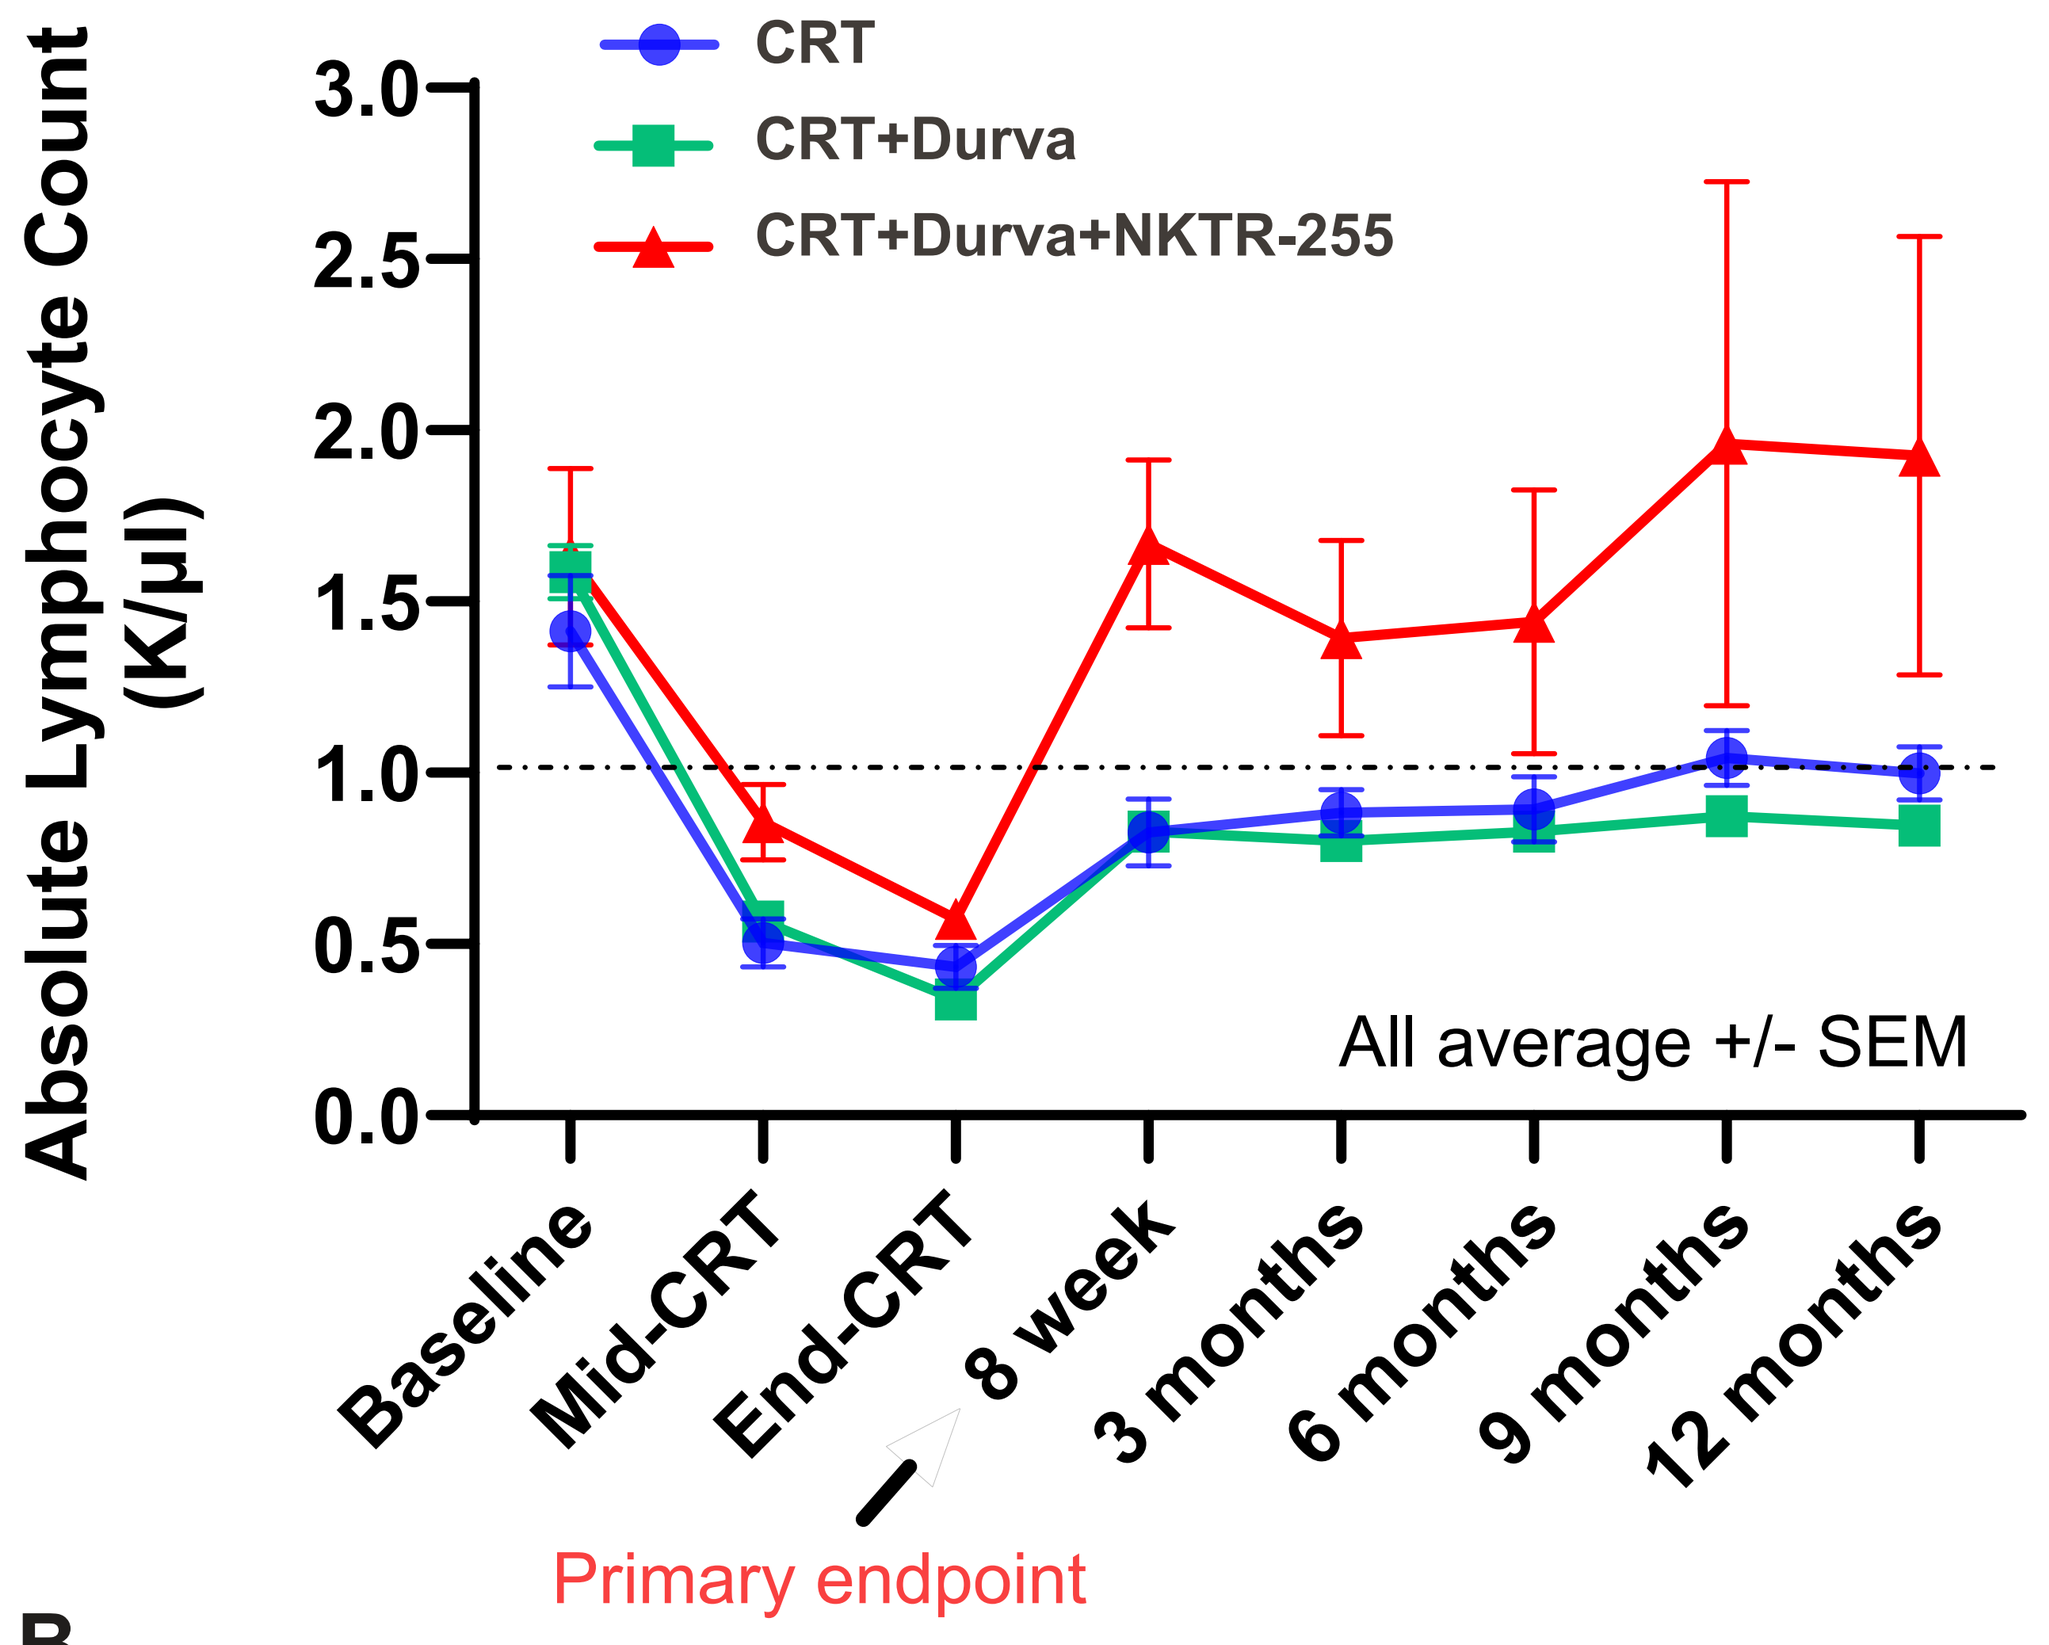
A.
Absolute Lymphocyte Count (K/µl)
CRT
CRT+Durva
CRT+Durva+NKTR-255
All average +/- SEM
Baseline
Mid-CRT
End-CRT
8 week
3 months
6 months
9 months
12 months
Primary endpoint

- Complete ALC normalization was met for 9 evaluable patients at 8 weeks post CRT
- At Cycle 1 day 8 (C1D8), the median increase in ALC was 2.35-fold (range 1.46-4.33) and at C2D8, it was 3.57-fold (range 1.84-8.09).
- **8-week ALC was significantly higher (p<0.001)**
- The non-contemporaneous control groups of CRT (n=39) or CRT + Durva (n=121) do not fully restore back to normal ALC count (≥ 1 K/µl), even after 1 year after CRT.

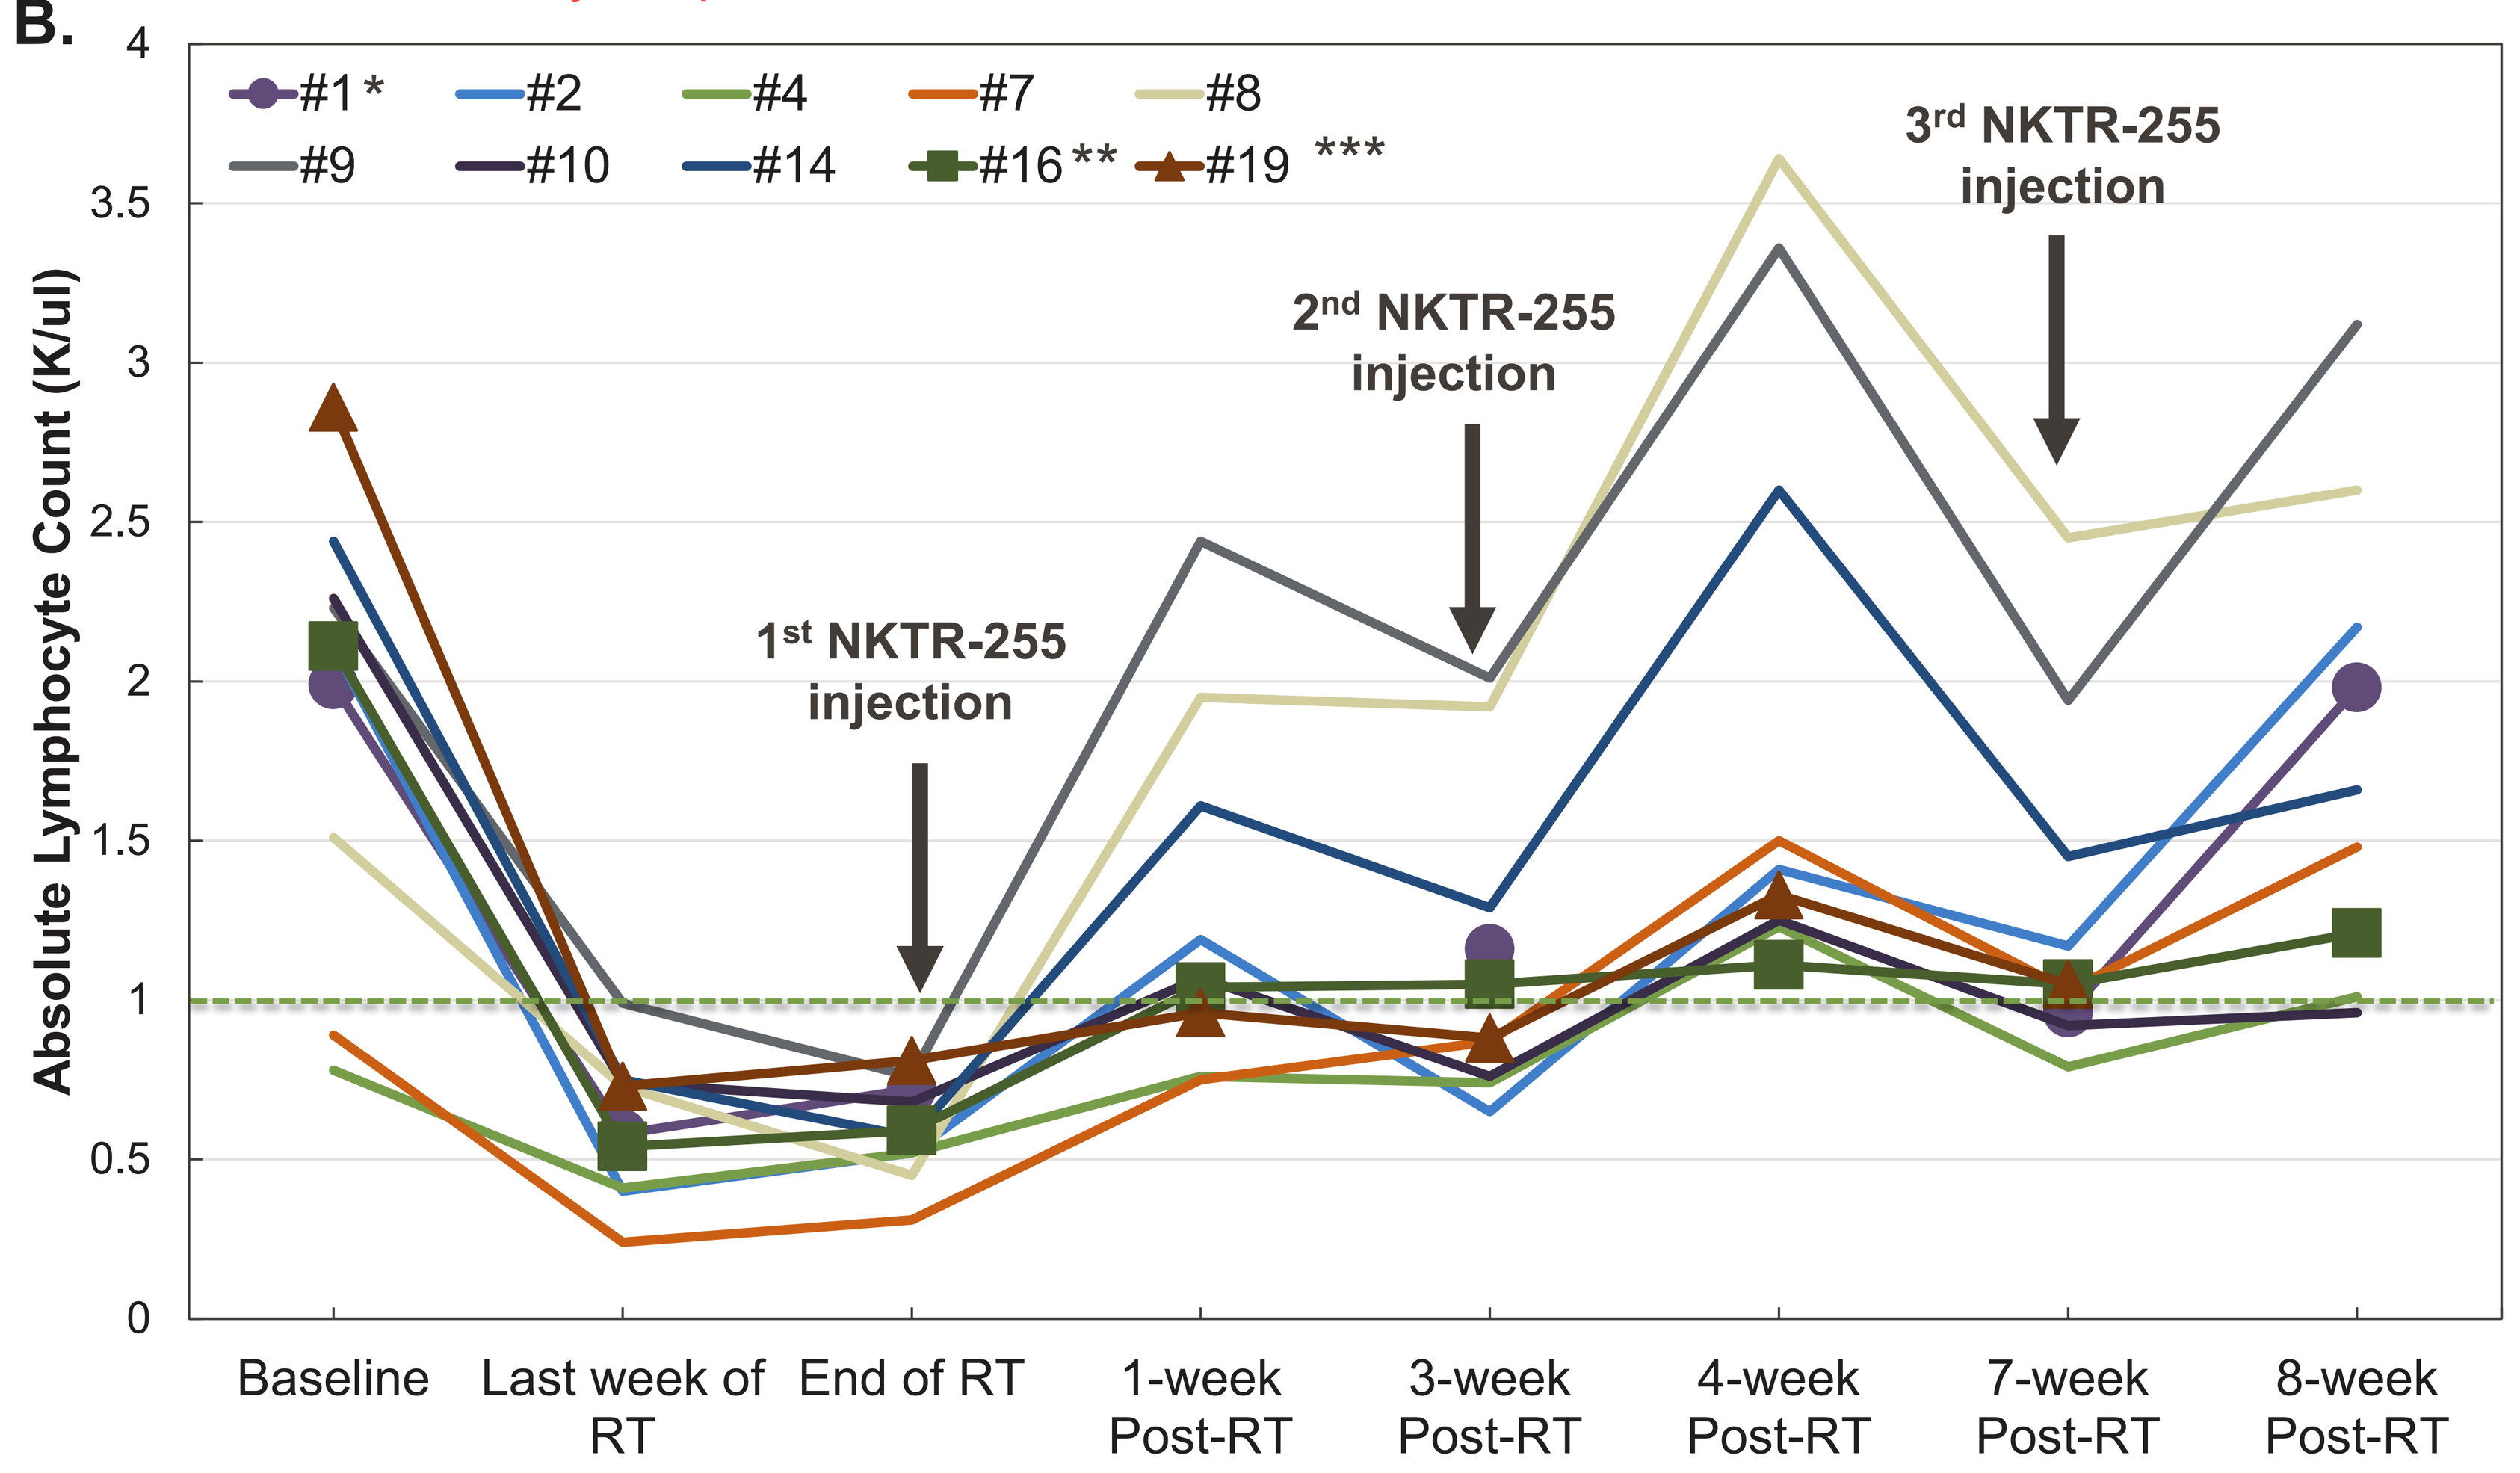
B.
#1 *
#2
#4
#7
#8
#9
#10
#14
#16 **
#19 ***
Absolute Lymphocyte Count (K/ul)
1st NKTR-255 injection
2nd NKTR-255 injection
3rd NKTR-255 injection
Baseline
Last week of RT
End of RT
1-week Post-RT
3-week Post-RT
4-week Post-RT
7-week Post-RT
8-week Post-RT

Fig. 3 Circulating lymphocyte counts following NKTR-255 injection (N=10). A. Lymphocyte count recovery trends in standard of care settings (CRT alone or CRT + Durva) and study treatment (CRT + NKTR-255 + Durva). For NKTR-255 containing-arm the samples counts were: 8 wk n=9; 3 mo n=4, 6 mo n=4; 9 mo n=2; 12 mo n=2. **B**. Lymphocyte count at 8-week post RT in 3 groups. C. Individual lymphocyte counts from baseline to 8-weeks post RT. *Patient #1 missed 1- and 4-week post-RT lab. **Patient #16 withdraw consent after 2 doses. ***Patient #19 had progressive disease after 2 doses and ALC at 8-week was not included. CRT = chemoradiation; Durva= Durvalumab; RT=radiation therapy.

NKTR-255 Pharmacokinetics

- Preliminary PK analyses showed no accumulation after repeat dosing.

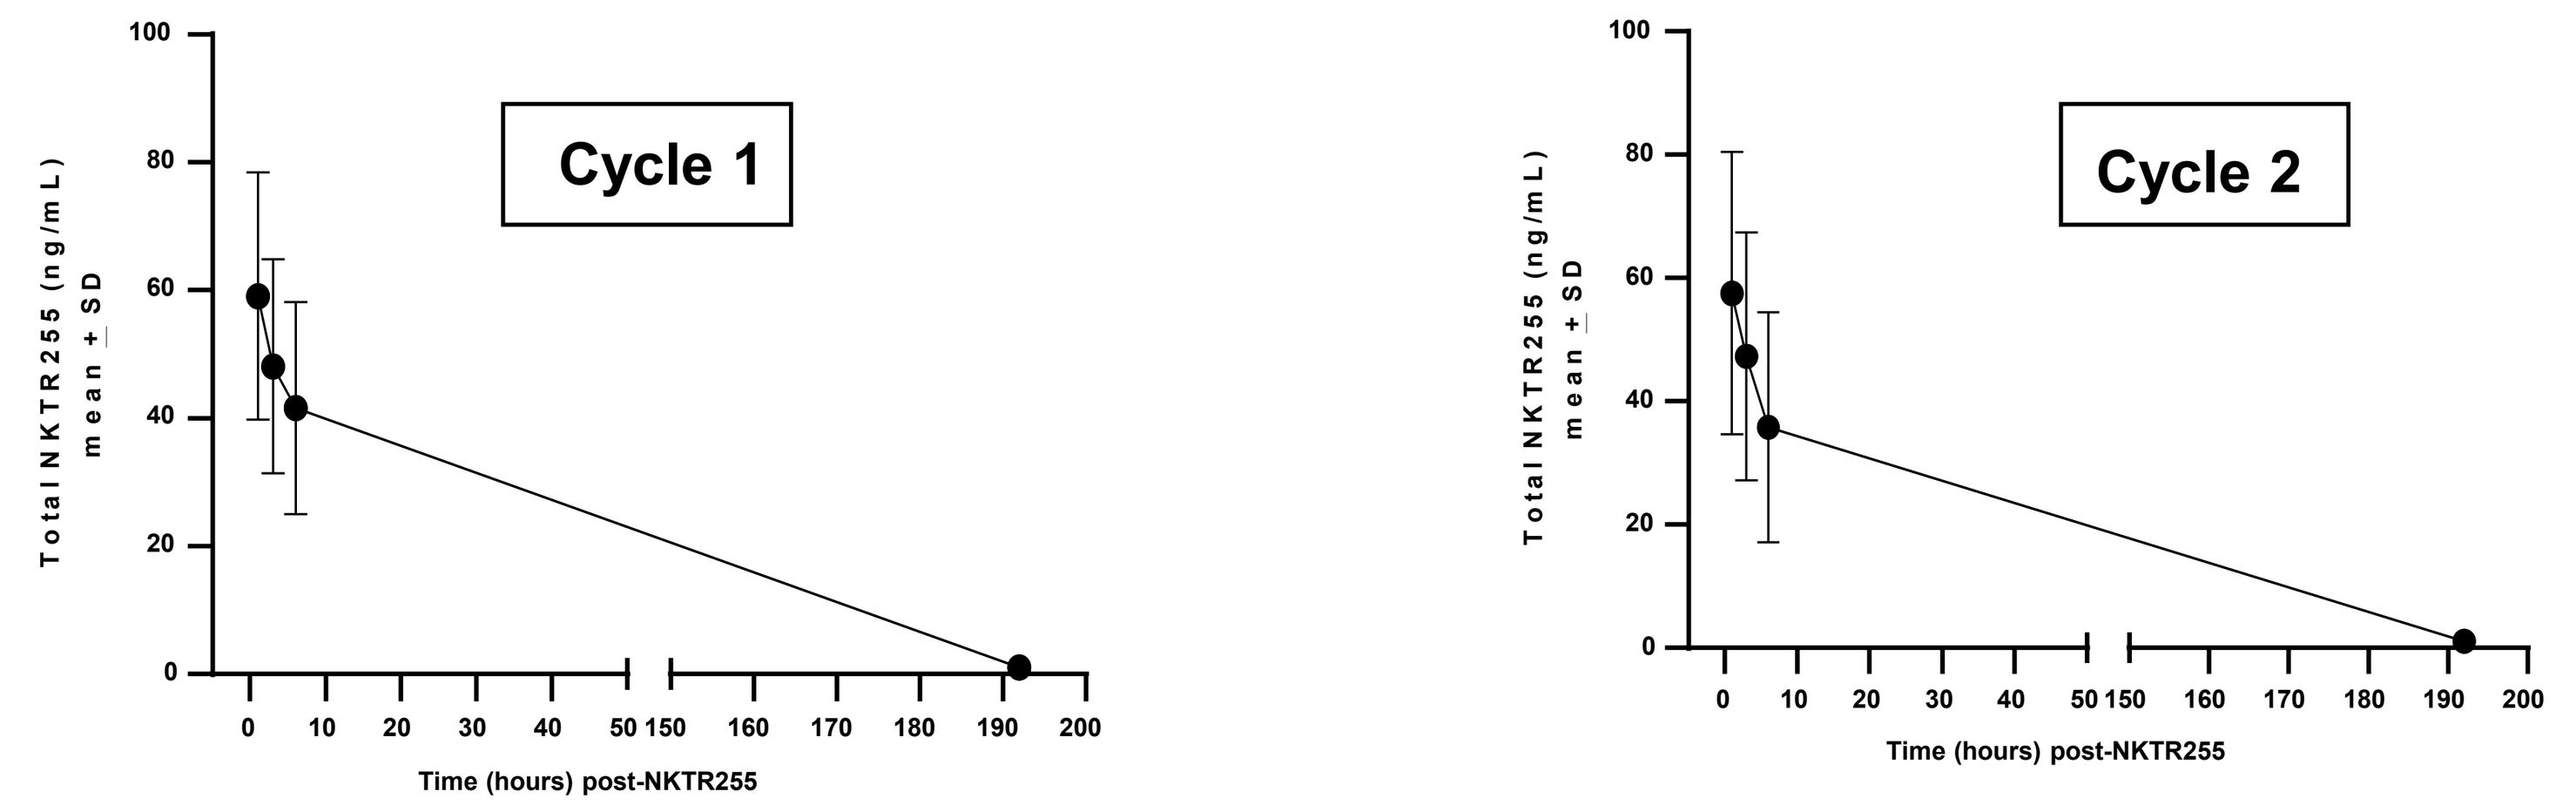
Cycle 1
Cycle 2
Total NKTR255 (ng/mL) mean ± SD
Time (hours) post-NKTR255

Fig. 4 Total NKTR-255 PK After Cycle 1 and 2 Administration. Data shown as NKTR-255 (ng/mL) ± SD. Cycle 1,n=11, Cycle 2, n=7. 3.0 µg/kg NKTR-255; PK=pharmacokinetic; SD=standard deviation.

NKTR-255 Enhances NK Cell Proliferation and Markers of Activation

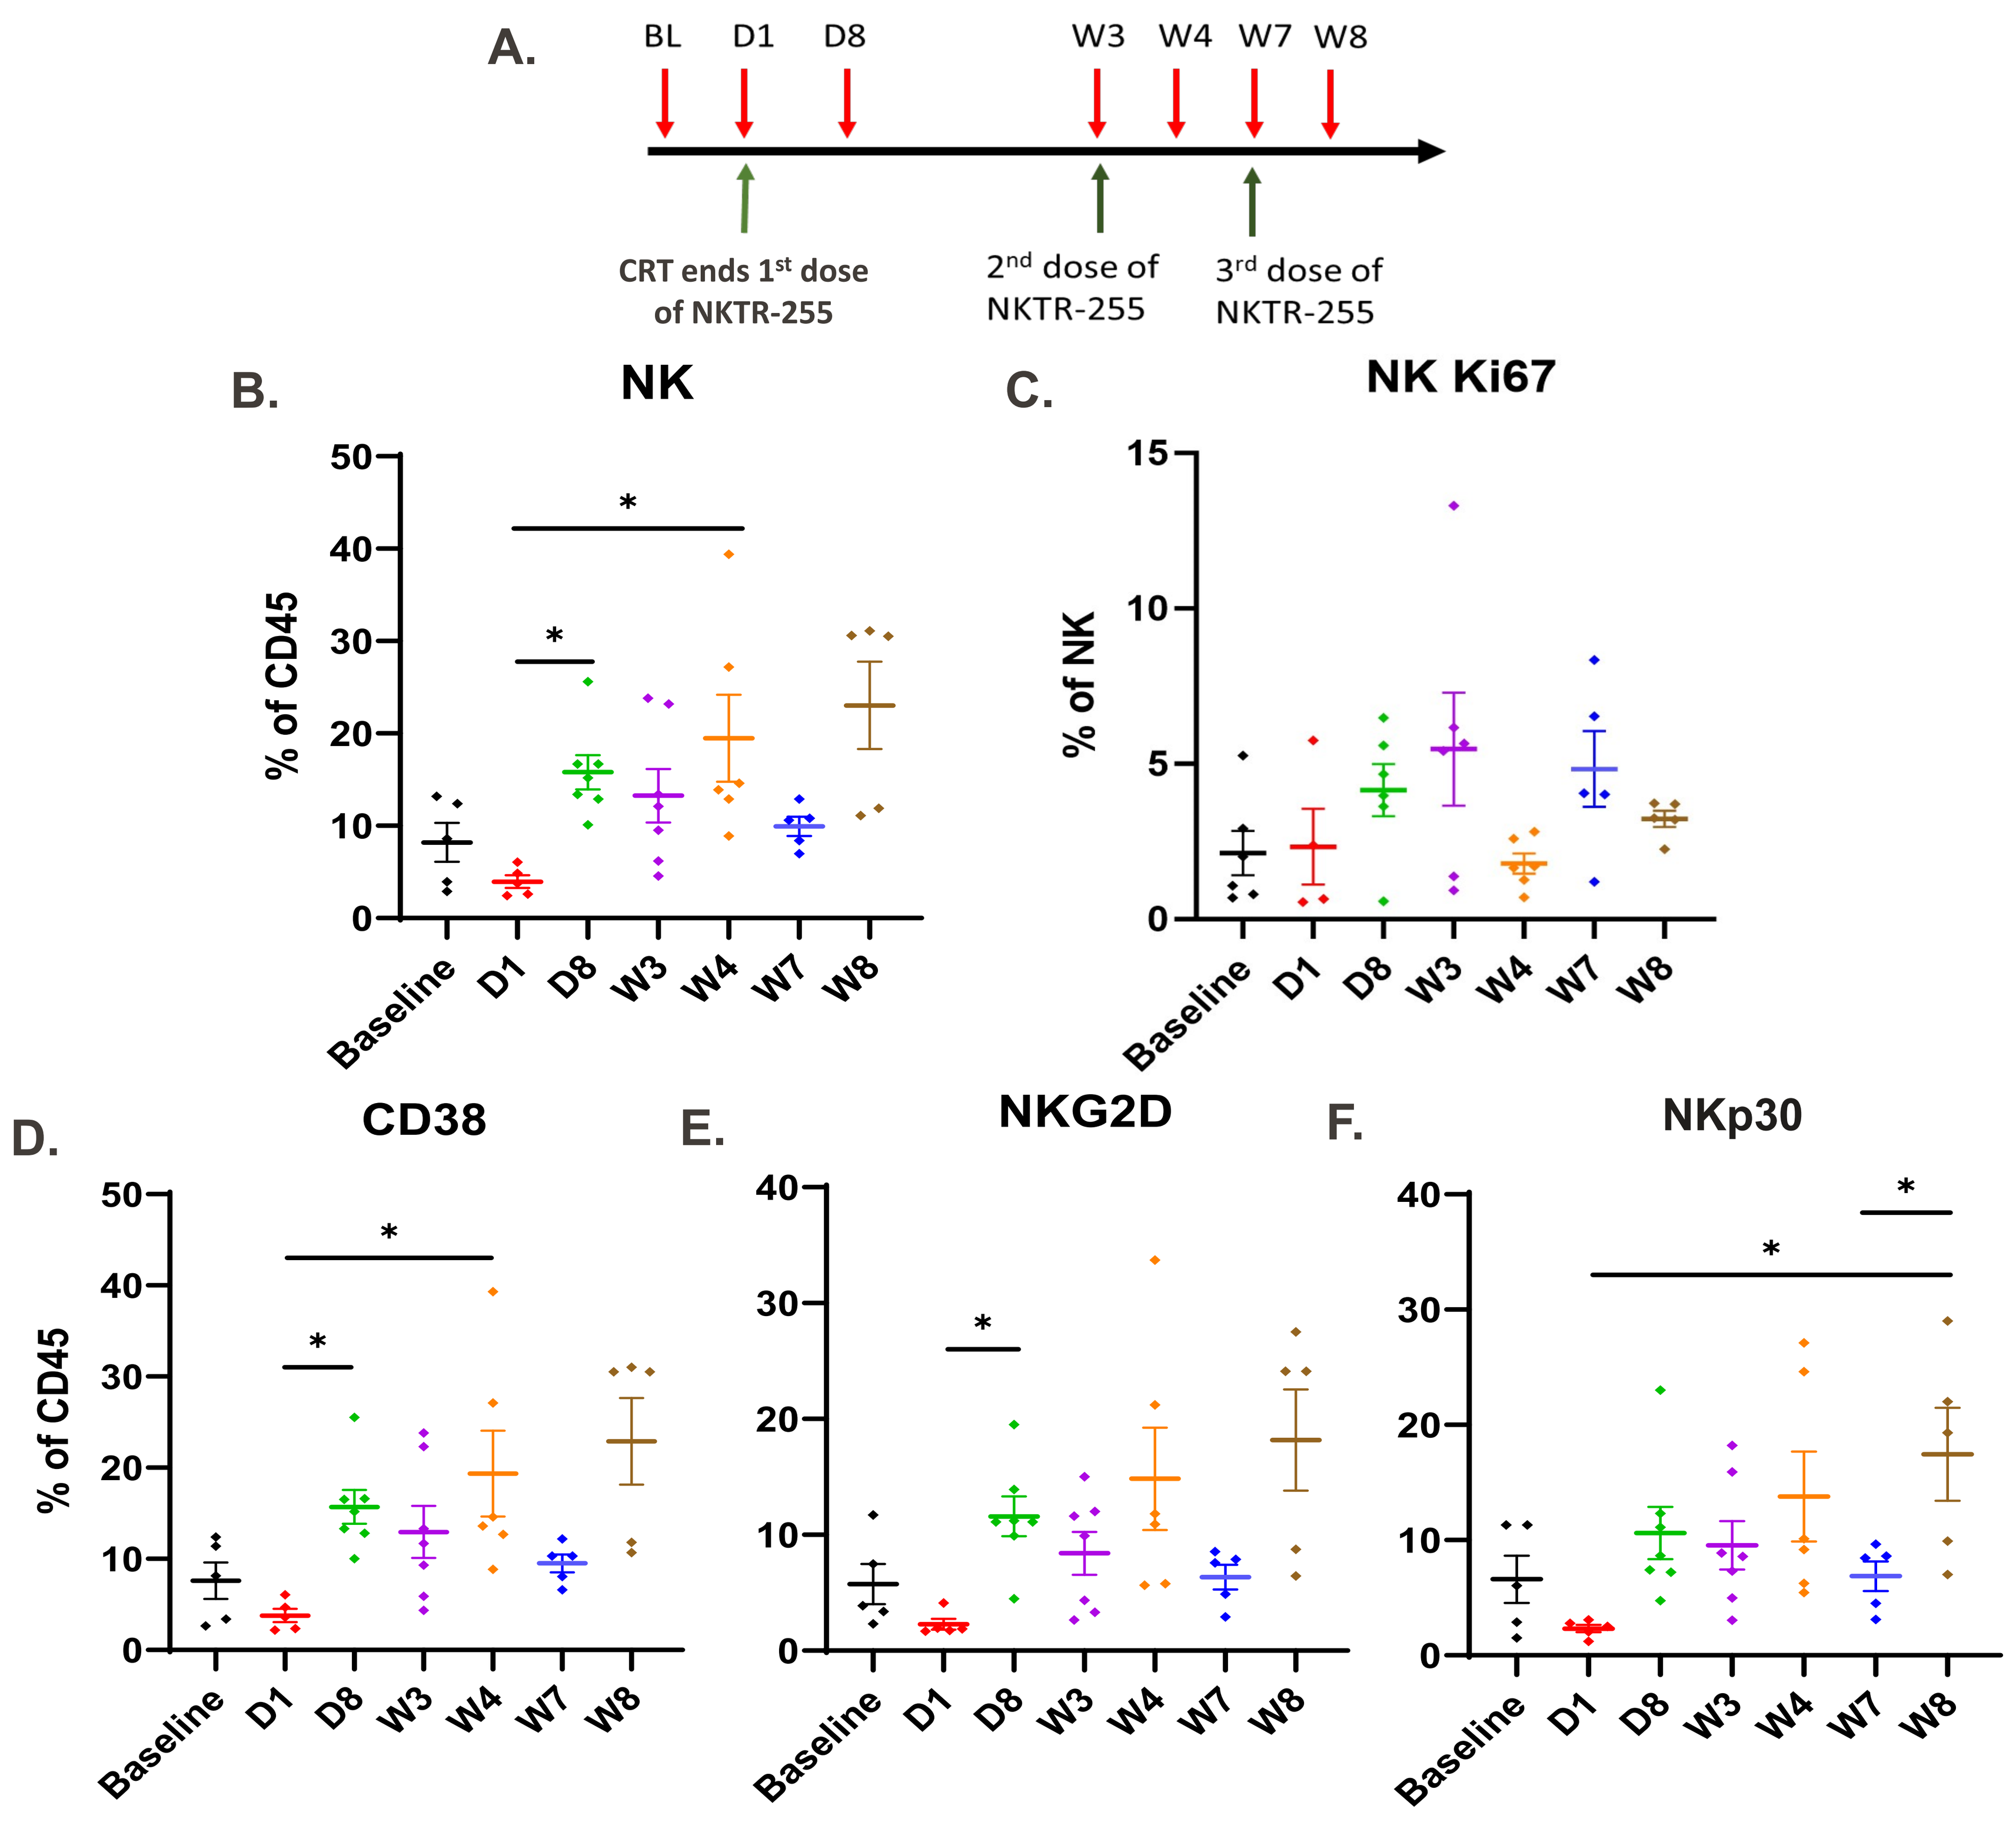
A.
BL
D1
D8
W3
W4
W7
W8
CRT ends 1st dose of NKTR-255
2nd dose of NKTR-255
3rd dose of NKTR-255
B.
NK
% of CD45
C.
NK Ki67
% of NK
D.
CD38
E.
NKG2D
F.
NKp30
Baseline
D1
D8
W3
W4
W7
W8

Fig. 5 Immune repertoire analysis of NK cells before and after NKTR-255. A. Schematic showing the experimental design where blood was drawn from seven subjects. **B.** Data points representing changes in NK cells before and after NKTR-255 injection at various time points. An increase in NK cell proliferation and activation markers **C.** Ki67, **D.** CD38, **E.** NKG2D, and **F**. NKp30 was observed following NKTR-255 injection across different time points. A mixed model analysis was conducted using SPSS 24. Data are reported as mean ± SE, with * indicating statistical significance (p < 0.05).

Conclusion

- The combination of NKTR-255 with CRT+Durva was safe and tolerable with a safety profile in-line with previous clinical reports .
- The NKTR-255 containing-arm effectively reversed CRT-induced lymphopenia.
- Interim PD results demonstrated statistically significant superiority of CRT+Durva with NKTR-255 versus non-contemporaneous CRT or CRT+Durva control groups from week 8 onward treated in the same setting.
- Additional pharmacodynamic findings following NKTR-255 administration show increased markers of NK cell activation, enhancing the anti-tumor activity after CRT.
- After reaching the initial safety and efficacy threshold, the trial continues to accrual at MDACC (NCT05632809).

References

1. Robinson, T. O., Hegde, S. M., Chang, A., Gangadharan, A., Rivas, S., Madakamutil, L., Zalevsky, J., Miyazaki, T., & Schluns, K. S. (2021). NKTR-255 is a polymer-conjugated IL-15 with unique mechanisms of action on T and natural killer cells. Journal of Clinical Investigation, 131(19). https://doi.org/10.1172/jci144365
2. Pilones, K. A., Charpentier, M., Garcia-Martinez, E., Daviaud, C., Kraynak, J., Aryankalayil, J., Formenti, S. C., & Demaria, S. (2020). Radiotherapy cooperates with IL15 to induce antitumor immune responses. Cancer Immunology Research, 8(8), 1054–1063. https://doi.org/10.1158/2326-6066.cir-19-0338
3. Jing, W., Xu, T., Wu, L., Lopez, P. B., Grassberger, C., Ellsworth, S. G., Mohan, R., Hobbs, B. P., Blumenschein, G. R., Tu, J., Altan, M., Lee, P., Liao, Z., & Lin, S. H. (2022). Severe radiation-induced lymphopenia attenuates the benefit of durvalumab after concurrent chemoradiotherapy for NSCLC. JTO Clinical and Research Reports, 3(9), 100391. https://doi.org/10.1016/j.jtocrr.2022.100391
4. Friedes, C., Chakrabarti, T., Olson, S., Prichett, L., Brahmer, J. R., Forde, P. M., Voong, R. K., Marrone, K. A., Lam, V. K., Hann, C. L., Broderick, S. R., Battafarano, R. J., Ha, J. S., Bush, E. L., Yang, S. C., Hales, R. K., & Feliciano, J. L. (2021). Association of severe lymphopenia and disease progression in unresectable locally advanced non-small cell lung cancer treated with definitive chemoradiation and immunotherapy. Lung Cancer, 154, 36–43. https://doi.org/10.1016/j.lungcan.2021.01.022